Patients with moderate-to-severe psoriasis do not believe clearance of their skin is a realistic treatment goal: results from the largest global psoriasis patient survey

Warren R.B.(1), Armstrong A.(2), Boehncke W-H.(3), Fernández-Peñas P.(4), Bewley A.(5), Romiti R.(6), Rajagopalan M.(7), Jarvis S.(8), Hillmann O.(9), Boyd E.(10), Sodha M.(11), Vaid B.(12), Huneault L.(13), Fox T.(14)

(1)The Dermatology Centre, Salford Royal NHS Foundation Trust, Manchester Academic Health Science Centre, The University of Manchester, United Kingdom, (2)Department of Dermatology, Keck School of Medicine at USC, University of Southern California, Los Angeles, United States, (3)Geneva University Hospital, Division of Dermatology and Venerology, Department of Internal Medicine Specialities, Geneva, Switzerland, (4)The University of Sydney, Westmead Hospital, Department of Dermatology, Westmead, Australia, (5)Whipps Cross University Hospital and The Royal London Hospital, Department of Dermatology, London, United Kingdom, (6)Department of Dermatology, University of São Paulo, Brazil, (7)Apollo Hospitals, Department of Dermatology, Chennai, India, (8)Richford Gate Medical Practice, London, United Kingdom, (9)EUROPSO, Düsseldorf, Germany, (10)National Psoriasis Foundation, VP Strategic Alliances and Clinical Affairs, Portland, United States of America, (11)GfK, Basel, Switzerland, (12)Novartis Pharma AG, Communications, Basel, Switzerland, (13)Novartis Pharma AG, Public Relations, Basel, Switzerland, (14)Novartis Pharma AG, Basel, Switzerland

Introduction

- Psoriasis is a chronic autoimmune disease that affects more than 125 million people worldwide
- Plaque psoriasis is the most common form of the disease and appears as raised red patches covered with a silvery white build-up of dead skin cells
- It has a significant physical and psychological impact on patients' lives as well as a number of significant associated comorbidities, including psoriatic arthritis, metabolic syndrome and depression[1-4]

The *Clear about Psoriasis* survey is the largest global survey of psoriasis patients to date. It offers robust data defining the impact of psoriasis on patients' daily lives – from their own perspective – and for the first time assesses what having clear skin means to them.

The survey aims to improve the understanding of patients' perspectives on clear skin and, importantly, the impact of not achieving it. The primary objective of the survey was to assess the current level of knowledge a patient has and their ability to achieve clear or almost clear skin.

Materials and methods

The survey, conducted between October 2015 and March 2016, is a global project supported by 25 patient groups from around the world.

Respondents (≥18 years old), recruited via patient organizations and online panels, came from 31 countries across Europe, North America, South America, Australia and Asia. Participants to the survey were anonymous and were told only that a survey on psoriasis was being conducted on behalf of a pharmaceutical company.

The 20-minute survey, which largely consisted of multiple choice questions, was conducted primarily online although some face-to-face interviews were conducted in selected countries where online research was deemed unfeasible.

The Self-Reported Psoriasis Area Severity Index (PASI)[5] was used to assess severity of the disease.*

Respondents were required to have plaque psoriasis for which they had received a medical diagnosis from a dermatologist and must not have participated in an online survey about psoriasis in the preceding four weeks. Only patients with moderate-to-severe plaque psoriasis were included in the analysis, defined as a PASI score ≥10, or 5.0–9.9 with plaques in visible and/or sensitive areas, such as the face or genitals[5].

*Three additional validated scoring methods were used to assess severity of the disease and health-related quality of life including the Medical Outcomes Study Sleep Scale (MOS-SS)[6], the World Health Organization-5 Quality of Life[7] and the Standford Presenteeism Scale[8]. These data will be shown at another congress.

Results

Participants

The *Clear about Psoriasis* survey is the largest global survey to date of people living with psoriasis, with 398,230 people invited to take part and responses from n=8,338 patients with moderate-to-severe plaque psoriasis.

Patient demographics			
Region			
Europe	59%	Australia	3%
North Amercia	23%	Asia	9%
South Amercia	6%		
Gender			
Male	45%		
Female	55%		
PASI score (average score 14.3)			
PASI 5.0–9.9	32%		
PASI ≥10	68%		
Relationship status			
Married	55%	In a relationship	16%
Single	25%	Other	4%
Occupation status			
Employed	66%	Student	4%
Unemployed	7%	Homemaker	7%
Retired	13%	Other	3%

Lack of adequate treatment

When asked about their current treatment, 57% of patients reported that they have not achieved clear or almost clear skin. Nearly a third of patients (28%) had to wait five years before receiving treatment that resulted in clear or almost clear skin.

Treatment expectations

While seven out of ten (70%) patients are comfortable talking to their healthcare professional about wanting clear or almost clear skin, 55% do not believe this is a realistic goal. Furthermore, during consultation only 41% of patients agreed clear or almost clear skin as a treatment target with their healthcare professional.

Of those who achieved clear or almost clear skin, 79% were comfortable telling their doctor they wanted to achieve this goal versus only 63% of patients who had not achieved clear or almost clear skin.

Impact on daily life

84% of participants reported having faced discrimination or humiliation.

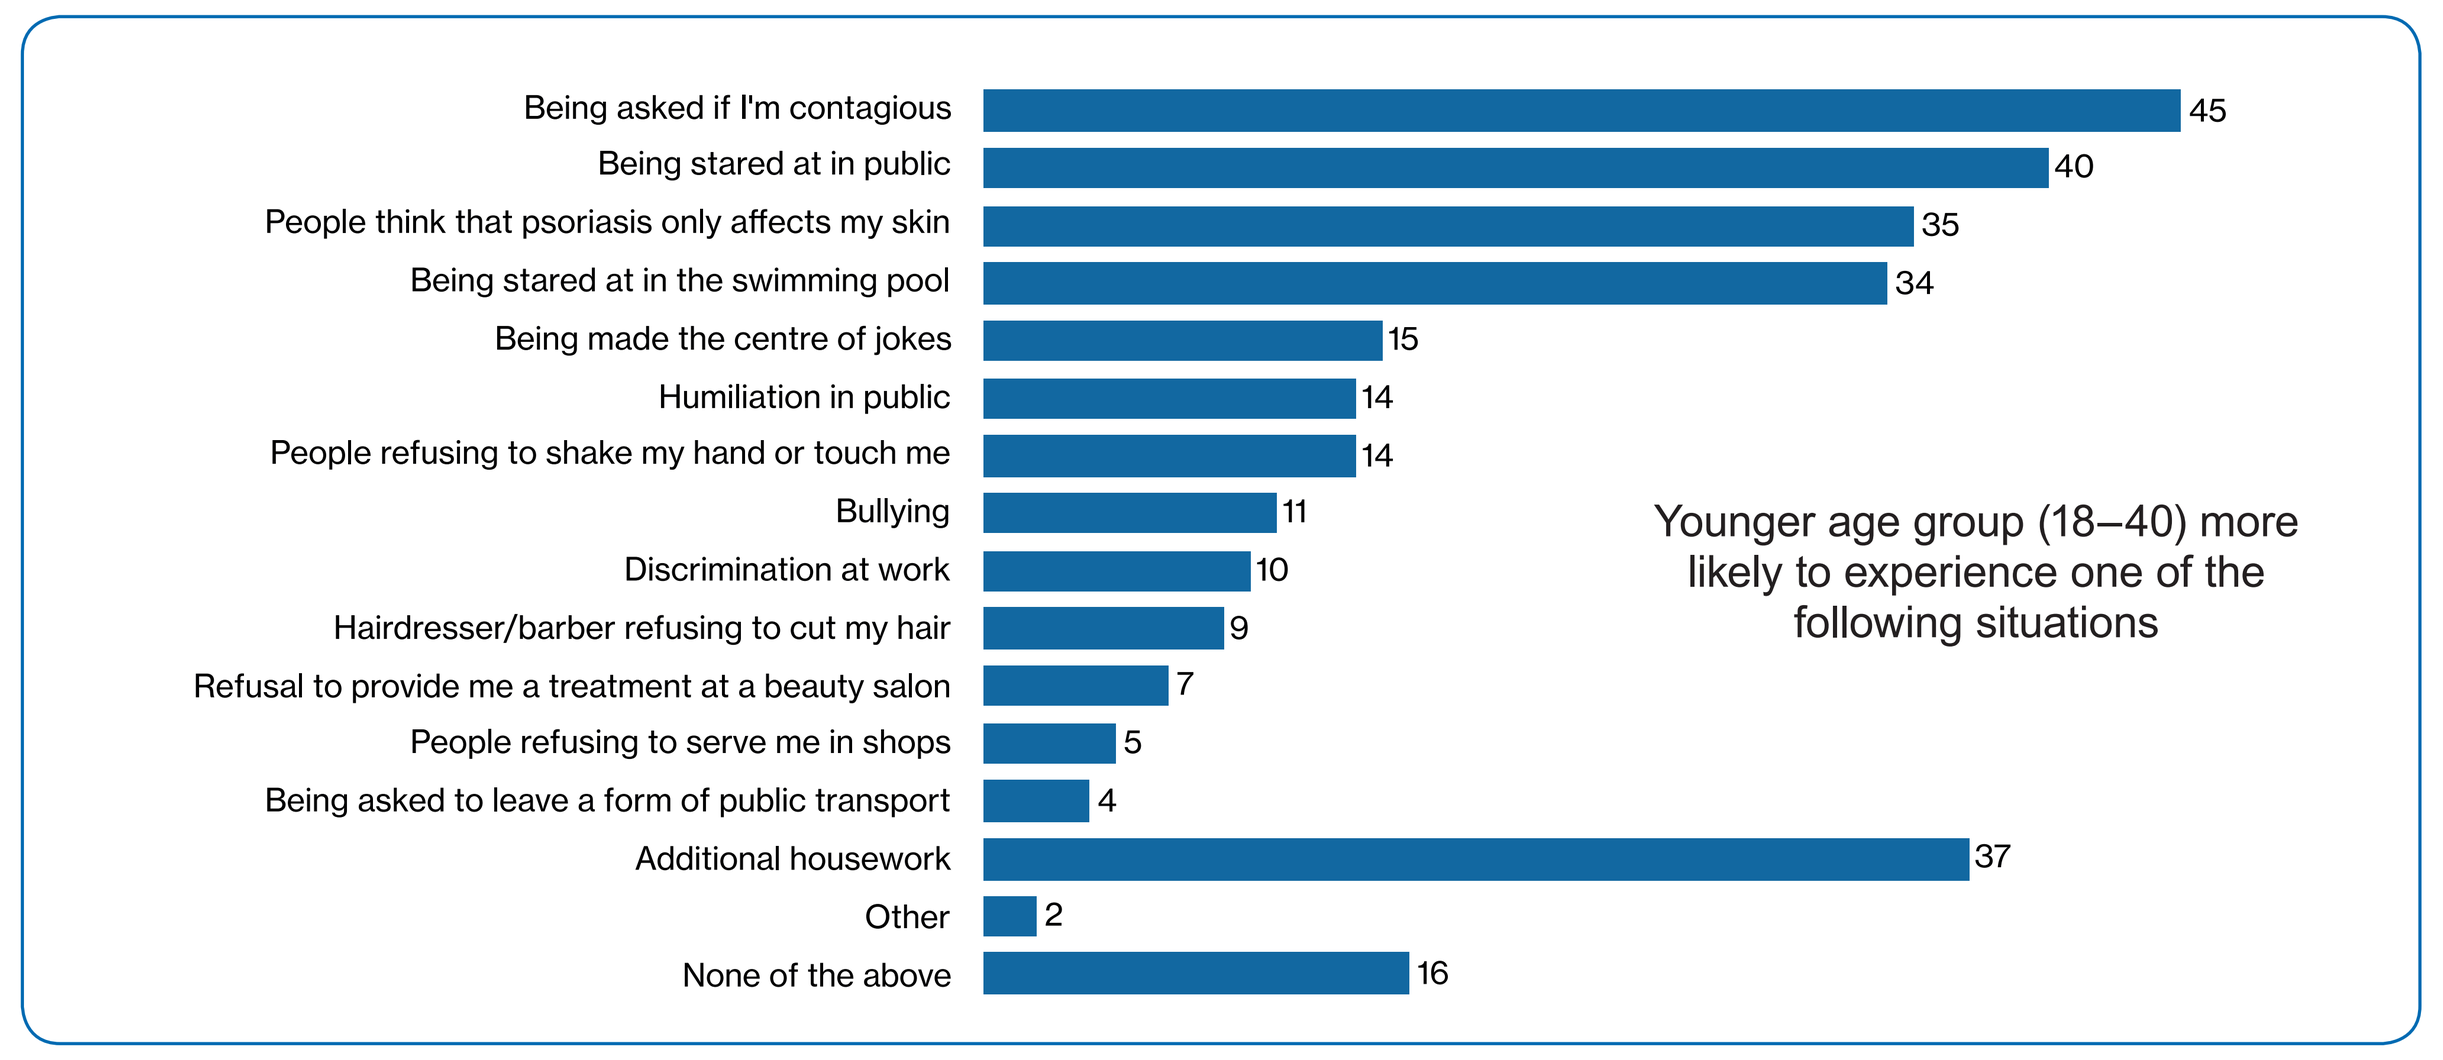
Being asked if I'm contagious 45
Being stared at in public 40
People think that psoriasis only affects my skin 35
Being stared at in the swimming pool 34
Being made the centre of jokes 15
Humiliation in public 14
People refusing to shake my hand or touch me 14
Bullying 11
Discrimination at work 10
Hairdresser/barber refusing to cut my hair 9
Refusal to provide me a treatment at a beauty salon 7
People refusing to serve me in shops 5
Being asked to leave a form of public transport 4
Additional housework 37
Other 2
None of the above 16
Younger age group (18–40) more likely to experience one of the following situations

Psoriasis and the reactions of others affect many aspects of patients' lives:

Mental health

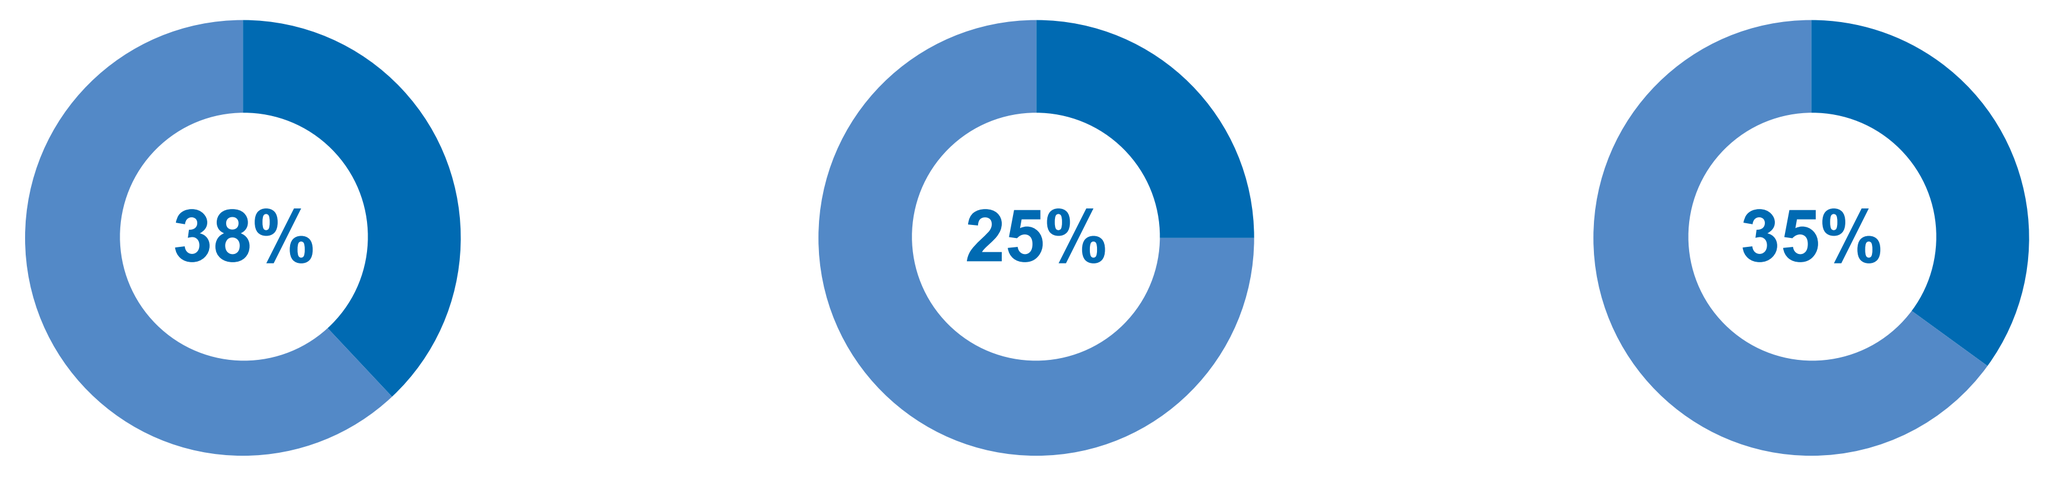
38%
25%
35%

have been diagnosed with a psychological condition as a result of their psoriasis

have been diagnosed with anxiety and **24%** with depression

reported that other people believe that psoriasis only affects their skin

Daily life

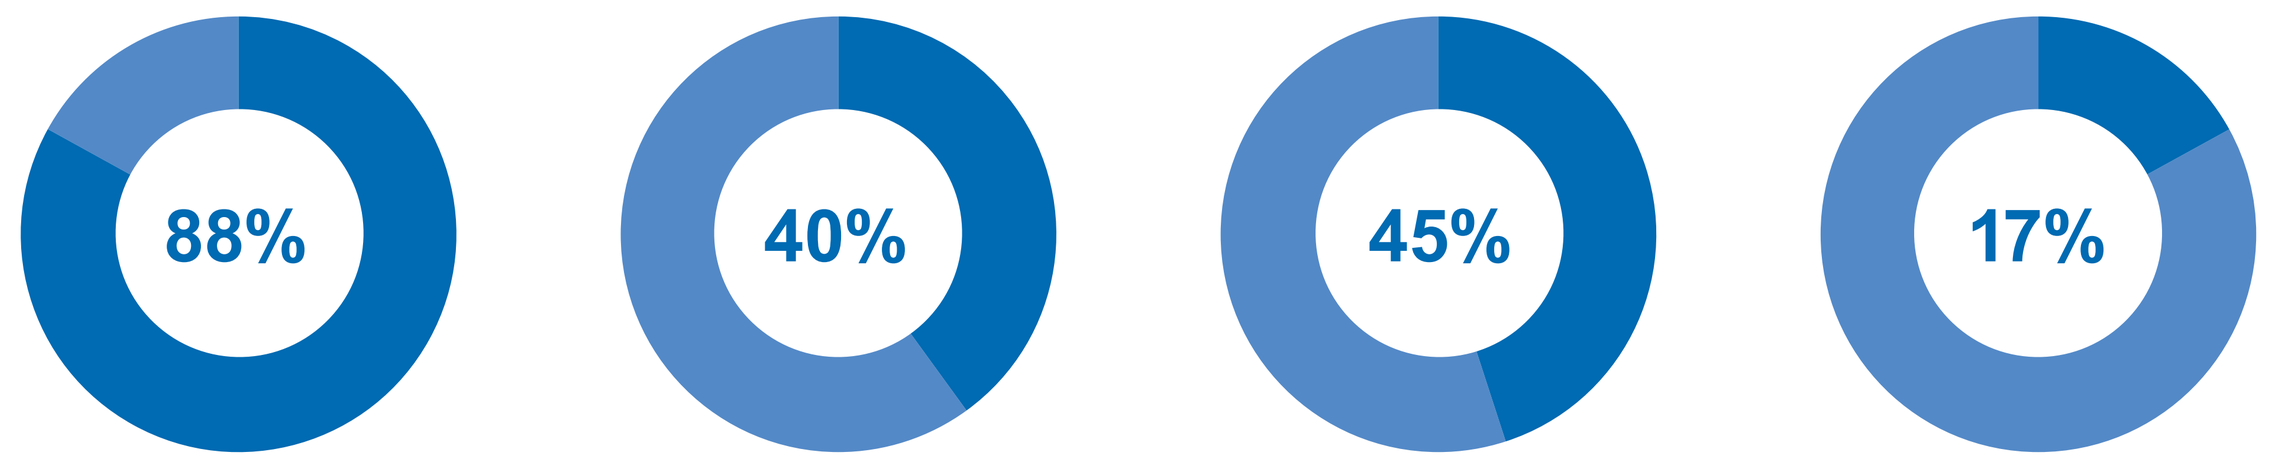
88%
40%
45%
17%

feel self-conscious or ashamed when doing activities

are being stared at in public

were asked if they were contagious

reported being turned away from consumer services such as beauticians, hair dressers, shops and public transport

Relationships

- **43%** of patients felt psoriasis had affected their relationships and made it difficult to form intimate relationships. Of these –

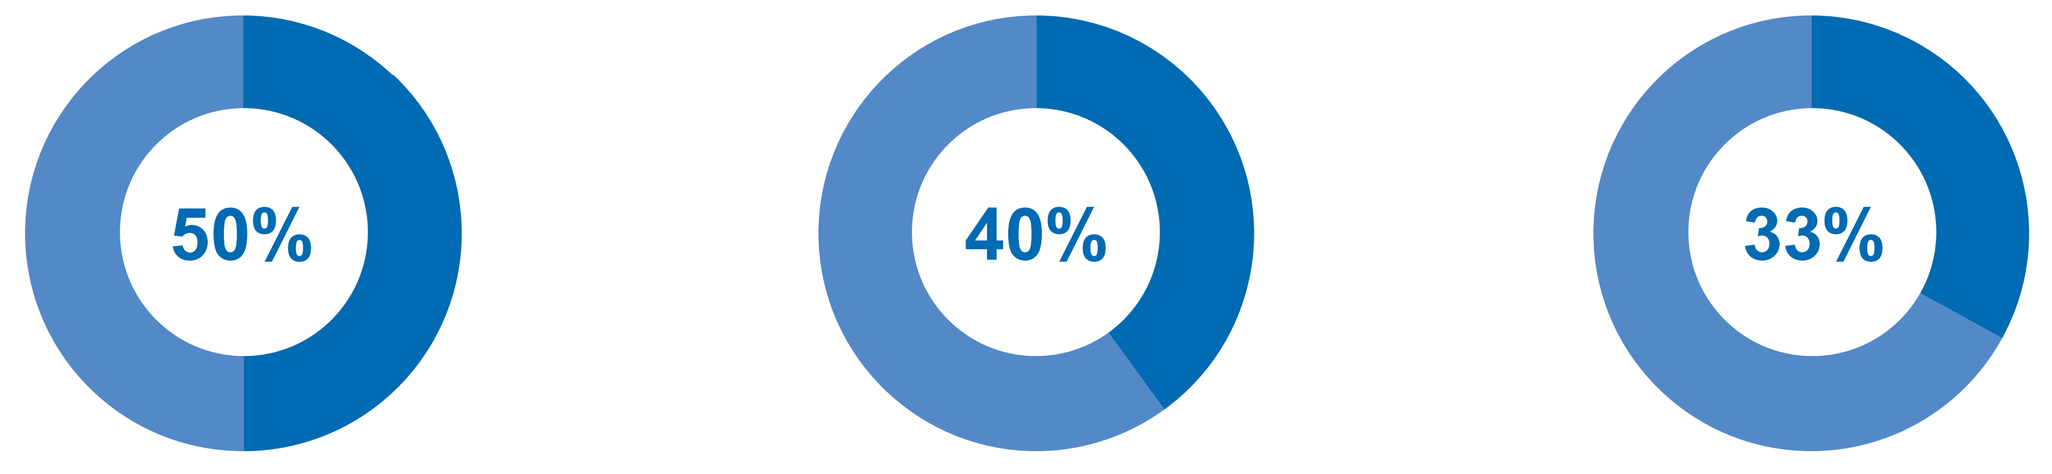
50%
40%
33%

avoid having sex because of their psoriasis

cannot stand the thought of someone seeing their skin (shedding)

feel inadequate as a spouse or partner

Working lives

- **41%** of patients missed at least one day of work in the last 6 months due to their psoriasis
- **54%** of patients felt psoriasis had an impact on their professional life and diminished their productivity and limited career opportunities. Of these –

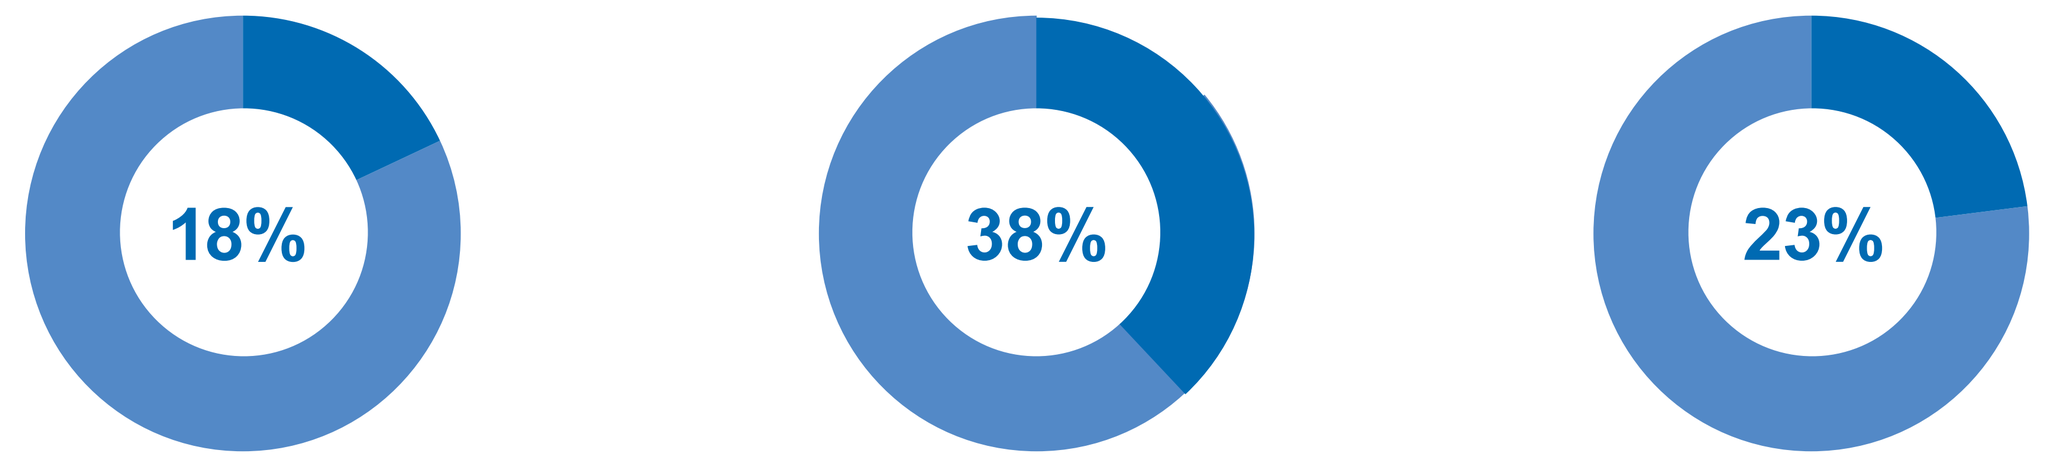
18%
38%
23%

worry they will lose their job

are not fully productive due to itching

are made fun of by people in their working environment

What does clear skin mean to patients?

The impact of psoriasis on patients' daily lives is highlighted by the activities they look forward to doing when they achieve clear or almost clear skin. The responses ranged from simple, everyday things such as lying on a beach or sunbathing (59%), going swimming (58%) and wearing black or dark clothes (40%). There are also more personal ones including having sexual relations with their partner (30%) or touching (24%) and hugging (23%) other people.

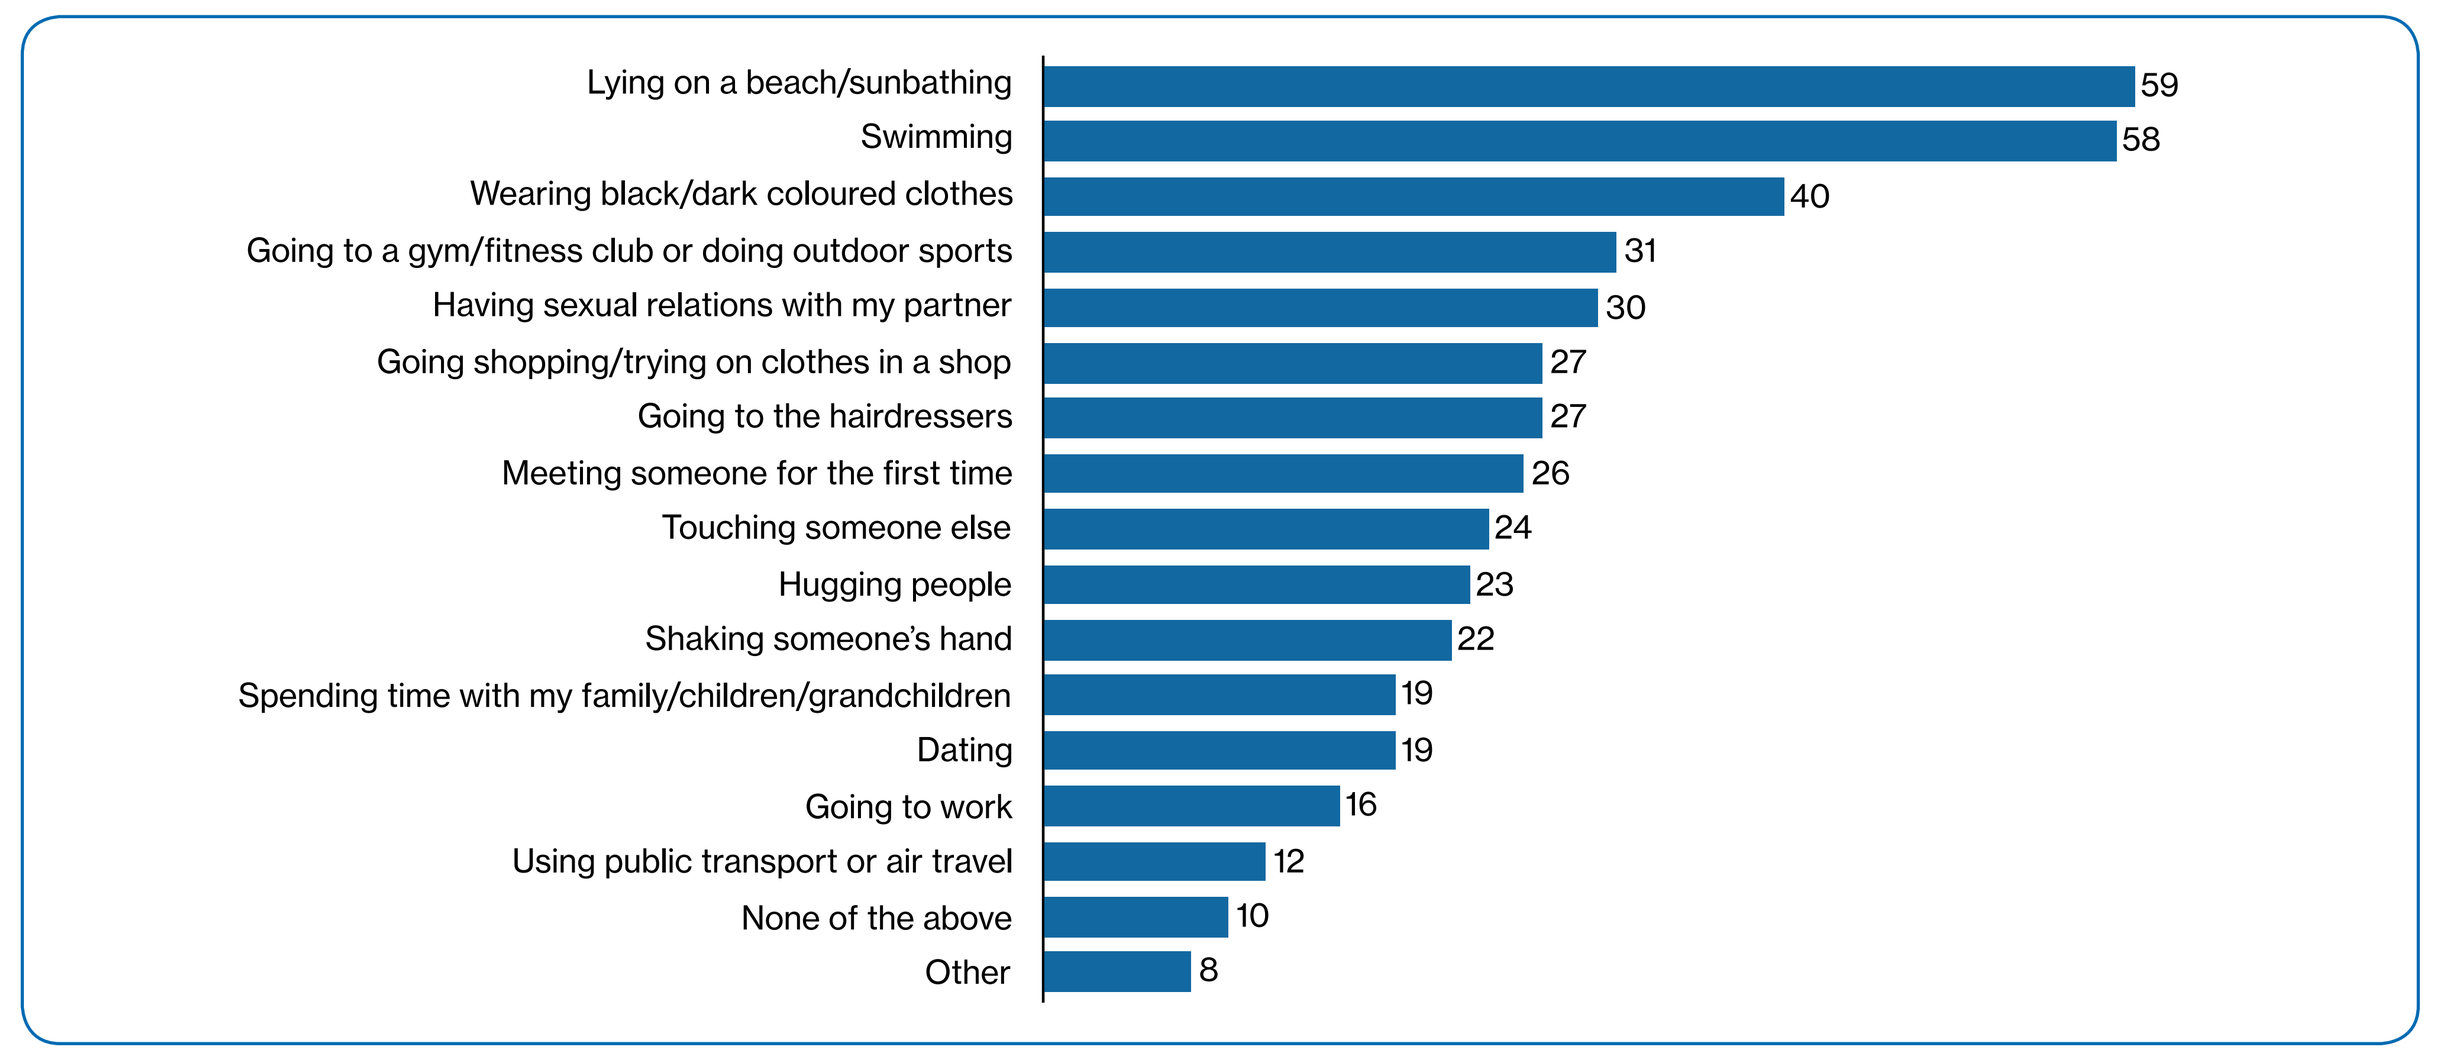
Lying on a beach/sunbathing 59
Swimming 58
Wearing black/dark coloured clothes 40
Going to a gym/fitness club or doing outdoor sports 31
Having sexual relations with my partner 30
Going shopping/trying on clothes in a shop 27
Going to the hairdressers 27
Meeting someone for the first time 26
Touching someone else 24
Hugging people 23
Shaking someone's hand 22
Spending time with my family/children/grandchildren 19
Dating 19
Going to work 16
Using public transport or air travel 12
None of the above 10
Other 8

Conclusions

- The *Clear about Psoriasis* survey is the largest global survey of psoriasis patients to date and the first to explore the perception of clear or almost clear skin and what it means to patients
- Inadequately managed psoriasis is associated with discrimination, humiliation and psychological conditions such as depression and anxiety, all of which can negatively impact a patient's day-to-day activities and quality of life
- Although the majority of patients are comfortable talking to their healthcare professional about wanting clear or almost clear skin, this is not regularly set as a primary treatment goal
 - > Patients who achieved clear or almost clear skin were more comfortable discussing this goal with their physicians than patients that had not achieved it
- Now clear or almost clear skin is more achievable than ever before this needs to be agreed by patients and their physicians as a treatment goal
- These data highlight a need for greater engagement and education of patients and healthcare professionals about clear or almost clear skin as an achievable treatment goal

References

1. International Federation of Psoriasis Associations (IFPA) World Psoriasis Day website. Available at: http://www.worldpsoriasisday.com. Accessed August 2016.
2. National Psoriasis Foundation. Psoriatic disease: about psoriasis. Available at: www.psoriasis.org/about-psoriasis. Accessed August 2016.
3. Mease PJ, Armstrong AW. Managing patients with psoriatic disease: the diagnosis and pharmacologic treatment of psoriatic arthritis in patients with psoriasis. *Drugs*. 2014;74:423–441.
4. Armstrong AW, Harskamp CT, Armstrong EJ. Psoriasis and metabolic syndrome: a systematic review and meta-analysis of observational studies. *J Am Acad Dermatol*. 2013;68(4):654–62.
5. Mrowietz U *et al*. Definition of treatment goals for moderate-to-severe psoriasis: a European consensus. *Arch Dermatol Res*. 2011;303(1):1–10.
6. Stewart AL *et al*. Conceptualization and measurement of health for adults in the Health Insurance Study. Vol II. Physical health in terms of functioning. *Santa Monica (CA): The RAND Corporation*. 1978:236–359.
7. Topp CW *et al*. The WHO-5 Well Being Index: a systematic review of the literature. *Psychother Psychosom*. 2015;84(3):167–76.
8. Koopman C *et al. J Occup Environ Med*. 2002;44(1):14–20.

DISCLOSURE

Novartis initiated and funded the study, which was conducted by the market research company Gesellschaft für Konsumforschung (GfK) Switzerland.